105

A STUDY OF THE SAFETY AND PHARMACOKINETICS OF MULTIPLE ASCENDING DOSES OF FV-100 IN HEALTHY SUBJECTS

Mark Matson[1], Helen Pentikis[2], Brian Boehlecke[3], Eric Wenzel[4], Geoff Henson[4], Amy Morris[4]
[1]Prism Research Inc., St. Paul, MN, USA/ [2]SAJE Consulting LLC, Baltimore, MD, USA/ [3]Rho Inc., Chapel Hill, NC, USA/ [4]Inhibitex, Inc., Alpharetta, GA, USA

Background

FV-100 is a 5'-valyl prodrug of CF-1743, a highly potent oral bicyclic nucleoside analogue, in development for treatment of herpes zoster, (shingles). *In vitro* studies demonstrated that CF-1743 rapidly enters varicella zoster infected cells and can completely stop viral replication within 5 minutes of exposure. This study assessed the safety and pharmacokinetics (PK) of multiple doses of FV-100 in healthy subjects aged 18-55 years.

Study Design / Methods

Healthy male and female subjects were randomized into sequential ascending dose cohorts: 100, 200, 400 and 800 mg given once daily for 7 days; a fifth cohort received 400 mg twice a day for 7 days. In each cohort, 6 subjects received FV-100 and 2 received placebo.

Plasma and urine were collected for 24 hours following the first dose and for 96 hours following the morning dose on Day 7 dosing. Plasma and urine were analyzed for FV-100 and CF-1743 by LC/MS/MS. A noncompartmental analysis was performed on the FV-100 and CF-1743 concentrations and PK parameters (C_{max}, AUC, t_{max}, $t_{1/2}$) were reported.

Subjects were followed for safety for 14 days after their last dose. Safety assessments included adverse event (AE) reports, physical exams (PE), electrocardiograms (ECG), and clinical laboratories.

Safety data from each cohort were reviewed when subjects completed the Study Day 22 evaluation, and advancement to the next dose level occurred only after pre-determined safety criteria were met.

Table 1. Summary of Adverse Events by Treatment Group and Severity

Severity	Placebo N = 12 n (%)	100 mg QD N = 6 n (%)	200 mg QD N = 6 n (%)	400 mg QD N = 6 n (%)	400 mg Q 12 Hours N = 6 n (%)	800 mg QD N = 6 n (%)
Grade 1	4 (33.3)	3 (50.0)	3 (50.0)	5 (83.3)	3 (50.0)	4 (66.7)
Grade 2	1 (8.3)	0 (0.0)	0 (0.0)	0 (0.0)	0 (0.0)	0 (0.0)

Table 2. Summary of Adverse Events in 2 or More Subjects by Treatment Group and Type

	Placebo N = 12 n (%)	100 mg QD N = 6 n (%)	200 mg QD N = 6 n (%)	400 mg QD N = 6 n (%)	400 mg Q 12 Hours N = 6 n (%)	800 mg QD N = 6 n (%)
Any AE	4 (33.3)	3 (50.0)	3 (50.0)	5 (83.3)	3 (50.0)	4 (66.7)
Headache	3 (25.0)	3 (50.0)	0	2 (33.3)	0	1 (16.7)
Nausea	0	0	1 (16.7)	1 (16.7)	1 (16.7)	1 (16.7)
Abdominal pain	2 (16.7)	0	0	0	0	0
Back pain	0	0	0	1 (16.7)	0	1 (16.7)
Dizziness	0	1 (16.7)	0	0	0	1 (16.7)

Figure 1. Pharmacokinetics of CF-1743 in Healthy Volunteers – Day 1

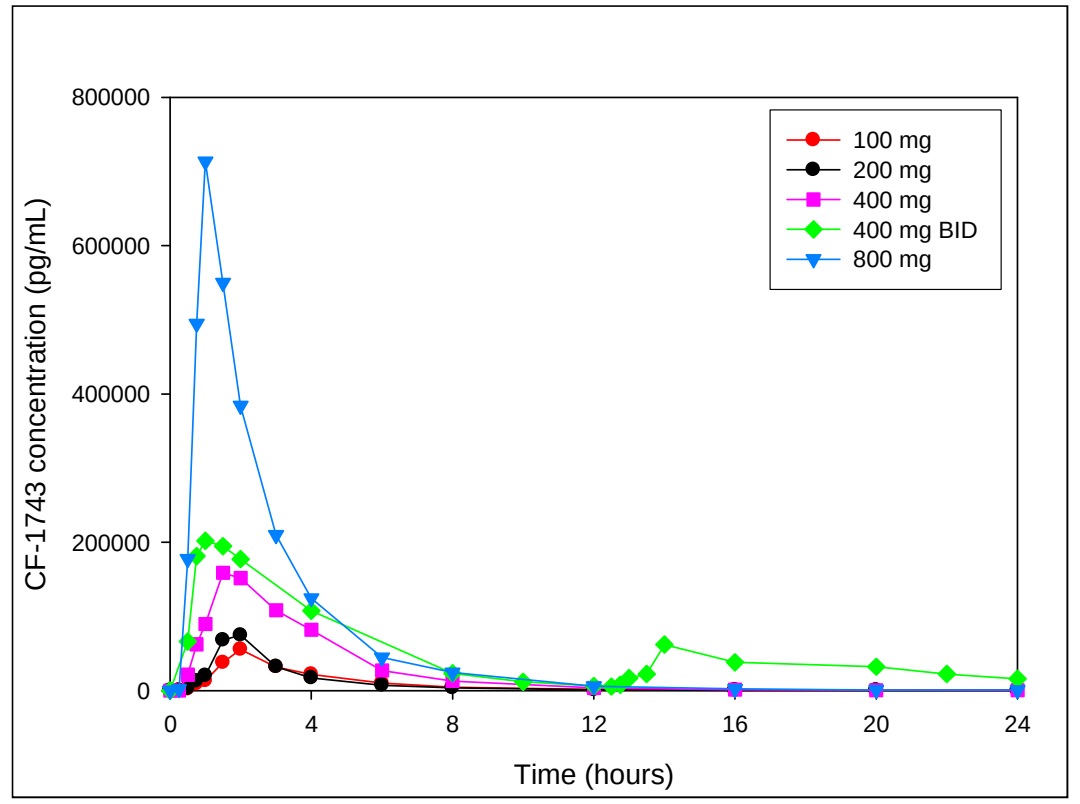
800000
600000
400000
200000
0
CF-1743 concentration (pg/mL)
0
4
8
12
16
20
24
Time (hours)
100 mg
200 mg
400 mg
400 mg BID
800 mg

Figure 2. Pharmacokinetics of CF-1743 in Healthy Volunteers – Day 7

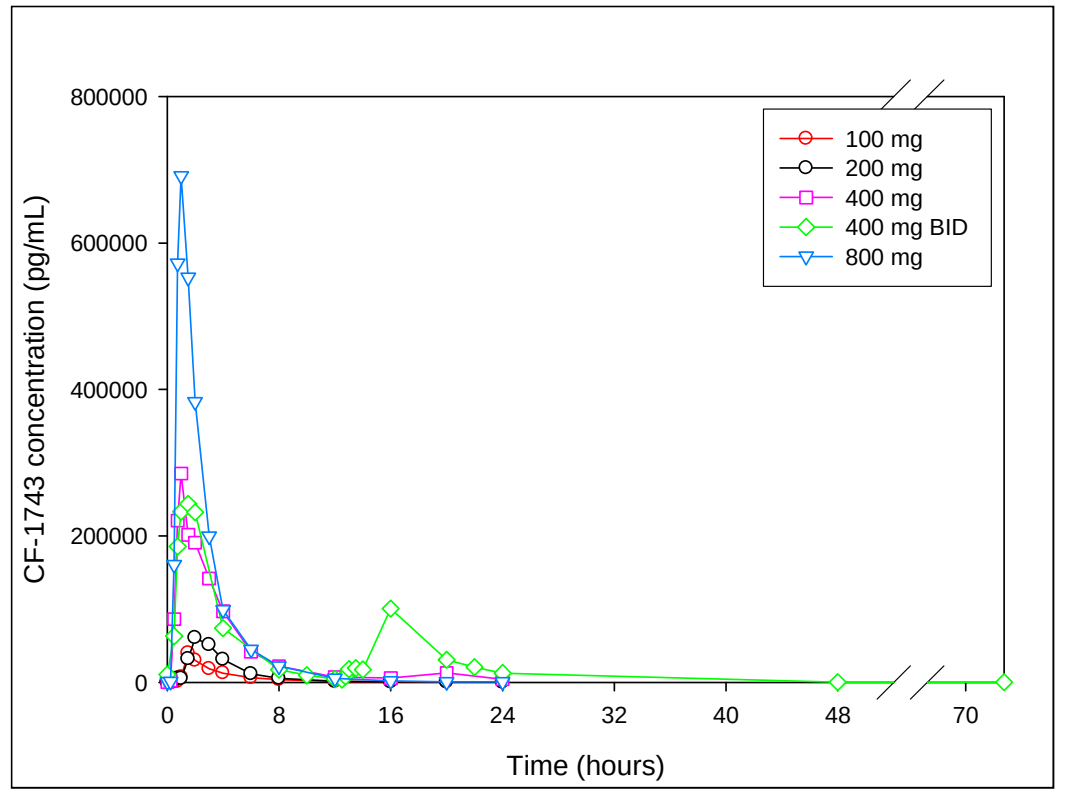
800000
600000
400000
200000
0
CF-1743 concentration (pg/mL)
0
8
16
24
32
40
48
70
Time (hours)
100 mg
200 mg
400 mg
400 mg BID
800 mg

Results

FV-100 concentrations were low for all dose cohorts. FV-100 was rapidly converted to CF-1743, the active metabolite, as evident by measurable CF-1743 plasma levels within 15 to 30 minutes of dosing. Both C_{max} and AUC for CF-1743 increased in a greater than dose proportional manner over the 100 mg to 800 mg dose range following single and multiple dose administration .

The observed accumulation of CF-1743, R_{obs}, was 0.8 to 1.8 and indicated that there was no appreciable accumulation following multiple dose administration. Renal clearance and percent of dose excreted was low for all dose cohorts, indicating that renal elimination is not likely to be an important pathway of elimination for FV-100 and CF-1743.

Twenty-two of 42 subjects reported at least 1 AEs (Table 2); 23 were thought by the Investigator to be possibly related to study drug. All AEs in subjects receiving FV-100 were mild and resolved prior to the end of the study. There were no clinically significant changes in PEs, vital signs, clinical laboratories or ECGs obtained throughout the trial.

Conclusions

- All doses of FV-100 were generally well tolerated; there were no trends for type, severity or frequency of AEs across dose levels.
- Concentrations for CF-1743 were above the EC_{50} value of 170 pg/mL for the entire 24 hour dosing interval for all dose cohorts following single and multiple dose administration, supporting a once a day regimen.
- Data from this study support moving into Phase II studies of FV-100 in patients with herpes zoster.

inhibitex®